A Phase 1b Study Evaluating IPI-926 in Combination with Gemcitabine in Patients with Metastatic Pancreatic Cancer

J. Stephenson[1], D. Richards[2], B. Wolpin[3], C. Becerra[4], J. Hamm[5], W. Messersmith[6], S. Devens[7], J. Cushing[7], J. Goddard[7], T. Schmalbach[7], C. S. Fuchs[3]

[1]US Oncology Research, Institute of Translational Oncology Research, Greenville, SC; [2]US Oncology Research, Tyler Cancer Center, Tyler, TX; [3]Dana Farber Cancer Institute, Boston, MA; [4]US Oncology Research, Texas Oncology, PA (North), Dallas, TX; [5]Norton Healthcare, Louisville, KY; [6]University of Colorado Hospital, Aurora, CO; [7]Infinity Pharmaceuticals, Inc., Cambridge, MA

Background

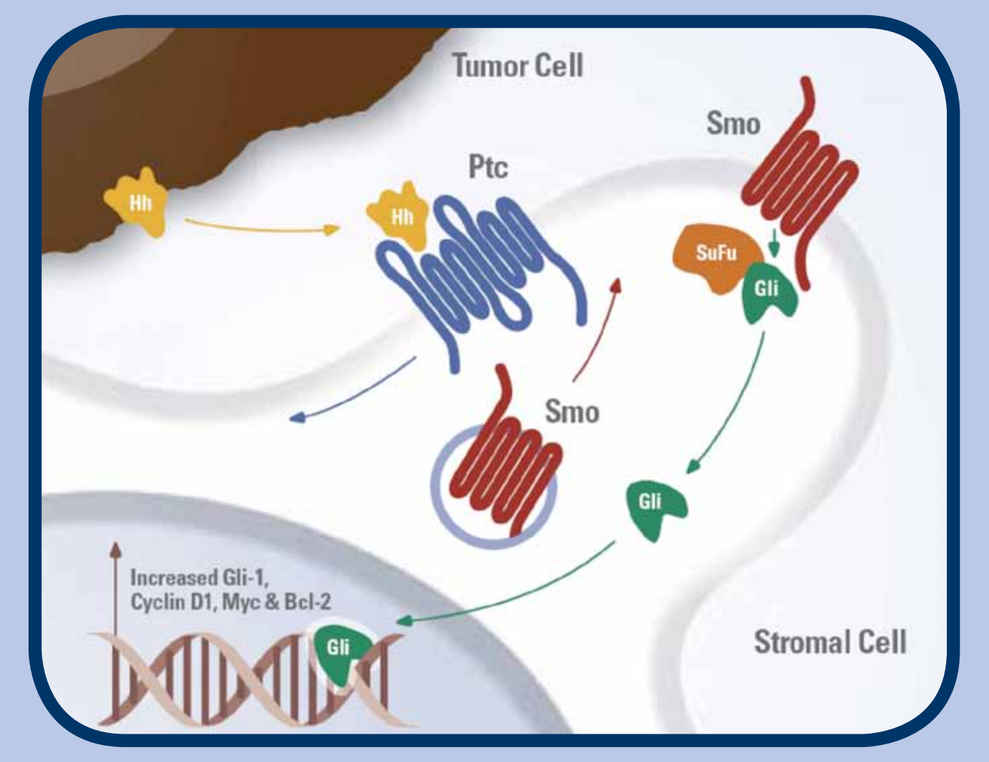
Tumor Cell
Smo
Ptc
Hh
Hh
SuFu
Gli
Smo
Gli
Increased Gli-1, Cyclin D1, Myc & Bcl-2
Gli
Stromal Cell

- In Hh ligand-dependent cancers, such as pancreatic cancer, Hh ligand produced by tumor cells acts through Patched (Ptc) on stromal cells in the tumor microenvironment. This mode of tumor-stroma paracrine signaling provides support for tumor growth and survival through mechanisms originating in the stromal cells.
- In pancreatic cancer models, inhibition of Hh signaling by antagonism of Smoothened (Smo) within the tumor microenvironment depletes the desmoplastic stroma, increases the vascularity of the tumor, and renders the tumor more accessible to chemotherapy.
- Pancreatic cancer is a devastating diagnosis with median progression-free survival of ~ 3 months and median overall survival of ~ 6 months. The overall response rate on gemcitabine is 5 – 10%, underscoring the urgent need for new therapies for this disease. (Moore, et al. J Clin Oncol 25:1960-6; Seitz et al. Oncology 18:43-7)

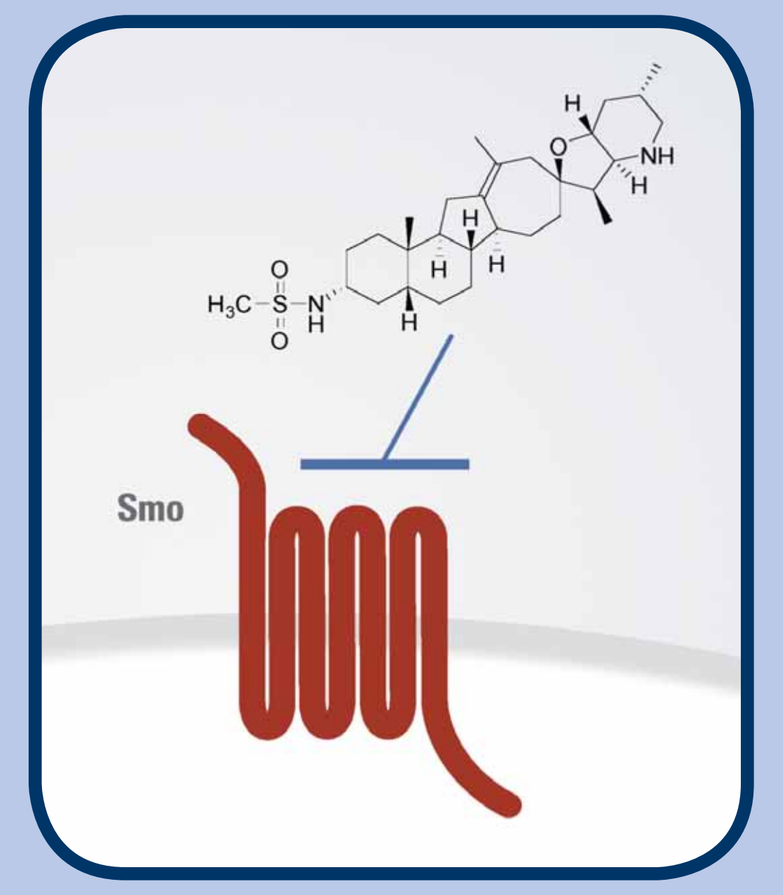
Smo

- IPI-926 is a potent, orally delivered small molecule that is currently being investigated as an inhibitor of Smo in multiple Phase 1 and Phase 2 trials.

Study Design

Phase 1b Objectives

- Determine the safety, maximum tolerated dose (MTD), and recommended Phase 2 dose for IPI-926 + gemcitabine
 - Evaluate pharmacokinetics of IPI-926, gemcitabine, and relevant metabolites

Study Design

- Phase 1b portion of a Phase 1b/2 study in which IPI-926 was dose-escalated with gemcitabine
- Cohorts of 3 or 6 patients were enrolled and administered daily, oral IPI-926 up to its single agent MTD (160 mg) plus gemcitabine (1000 mg/m² given 3 weeks on, 1 week off)
- Tolerability was determined during the first 28-day cycle

Patient Population

- Adults with pathologically confirmed diagnosis of metastatic pancreatic adenocarcinoma and at least 1 radiologically evaluable metastatic lesion (RECIST 1.1)
- ECOG Performance Score (0 or 1)
- No prior treatment with chemotherapy for pancreatic cancer and no known central nervous system metastases

Demographics and Disposition

Dose Group	Patients Treated (n)
110 mg	3
130 mg	6
160 mg	7

- Demographics (N = 16)
 - 14 males; 2 females
 - Mean age 69 years (range 58 - 86 years)
 - 15 White; 1 Black/African-American
- 5 patients currently on study
- Reasons for Discontinuation
 - Disease progression: 7 patients
 - Withdrawal of consent: 1 patient
 - Adverse events (gemcitabine toxicity): 1 patient, microangiopathic hemolytic anemia
 - Physician decision: 2 patients

Safety

IPI-926-Related TEAEs in ≥ 2 Patients	110 mg (N = 3)	130 mg (N = 6)	160 mg (N = 7)	Total (N = 16)
Fatigue				
Grade 1-2	3 (100)	3 (50)	2 (29)	8 (50)
Grade 3	0 (0)	0 (0)	1 (14)	1 (6)
Nausea				
Grade 1-2	1 (33)	4 (67)	1 (14)	6 (38)
Transaminases increased				
Grade 2	0 (0)	1 (17)	3 (43)	4 (25)
Grade 3	0 (0)	1 (17)	1 (14)	2 (13)
Vomiting				
Grade 1	0 (0)	4 (67)	0 (0)	4 (25)
Decreased appetite				
Grade 1-2	1 (33)	1 (17)	1 (14)	3 (19)
Anemia				
Grade 2	1 (33)	1 (17)	0 (0)	2 (13)
Diarrhea				
Grade 1	0 (0)	1 (17)	1 (14)	2 (13)
Edema peripheral				
Grade 1-2	0 (0)	0 (0)	2 (29)	2 (13)
Thrombocytopenia				
Grade 2	1 (33)	1 (17)	0 (0)	2 (13)

TEAE = treatment-emergent adverse event

- No deaths or TEAEs > Grade 3 related to IPI-926 or gemcitabine
- One DLT (130 mg): Grade 3 asymptomatic AST increase that resolved after IPI-926 dose reduction
- 10 (63%) patients required gemcitabine dose reduction (majority due to thrombocytopenia)

Tumor Marker CA 19-9

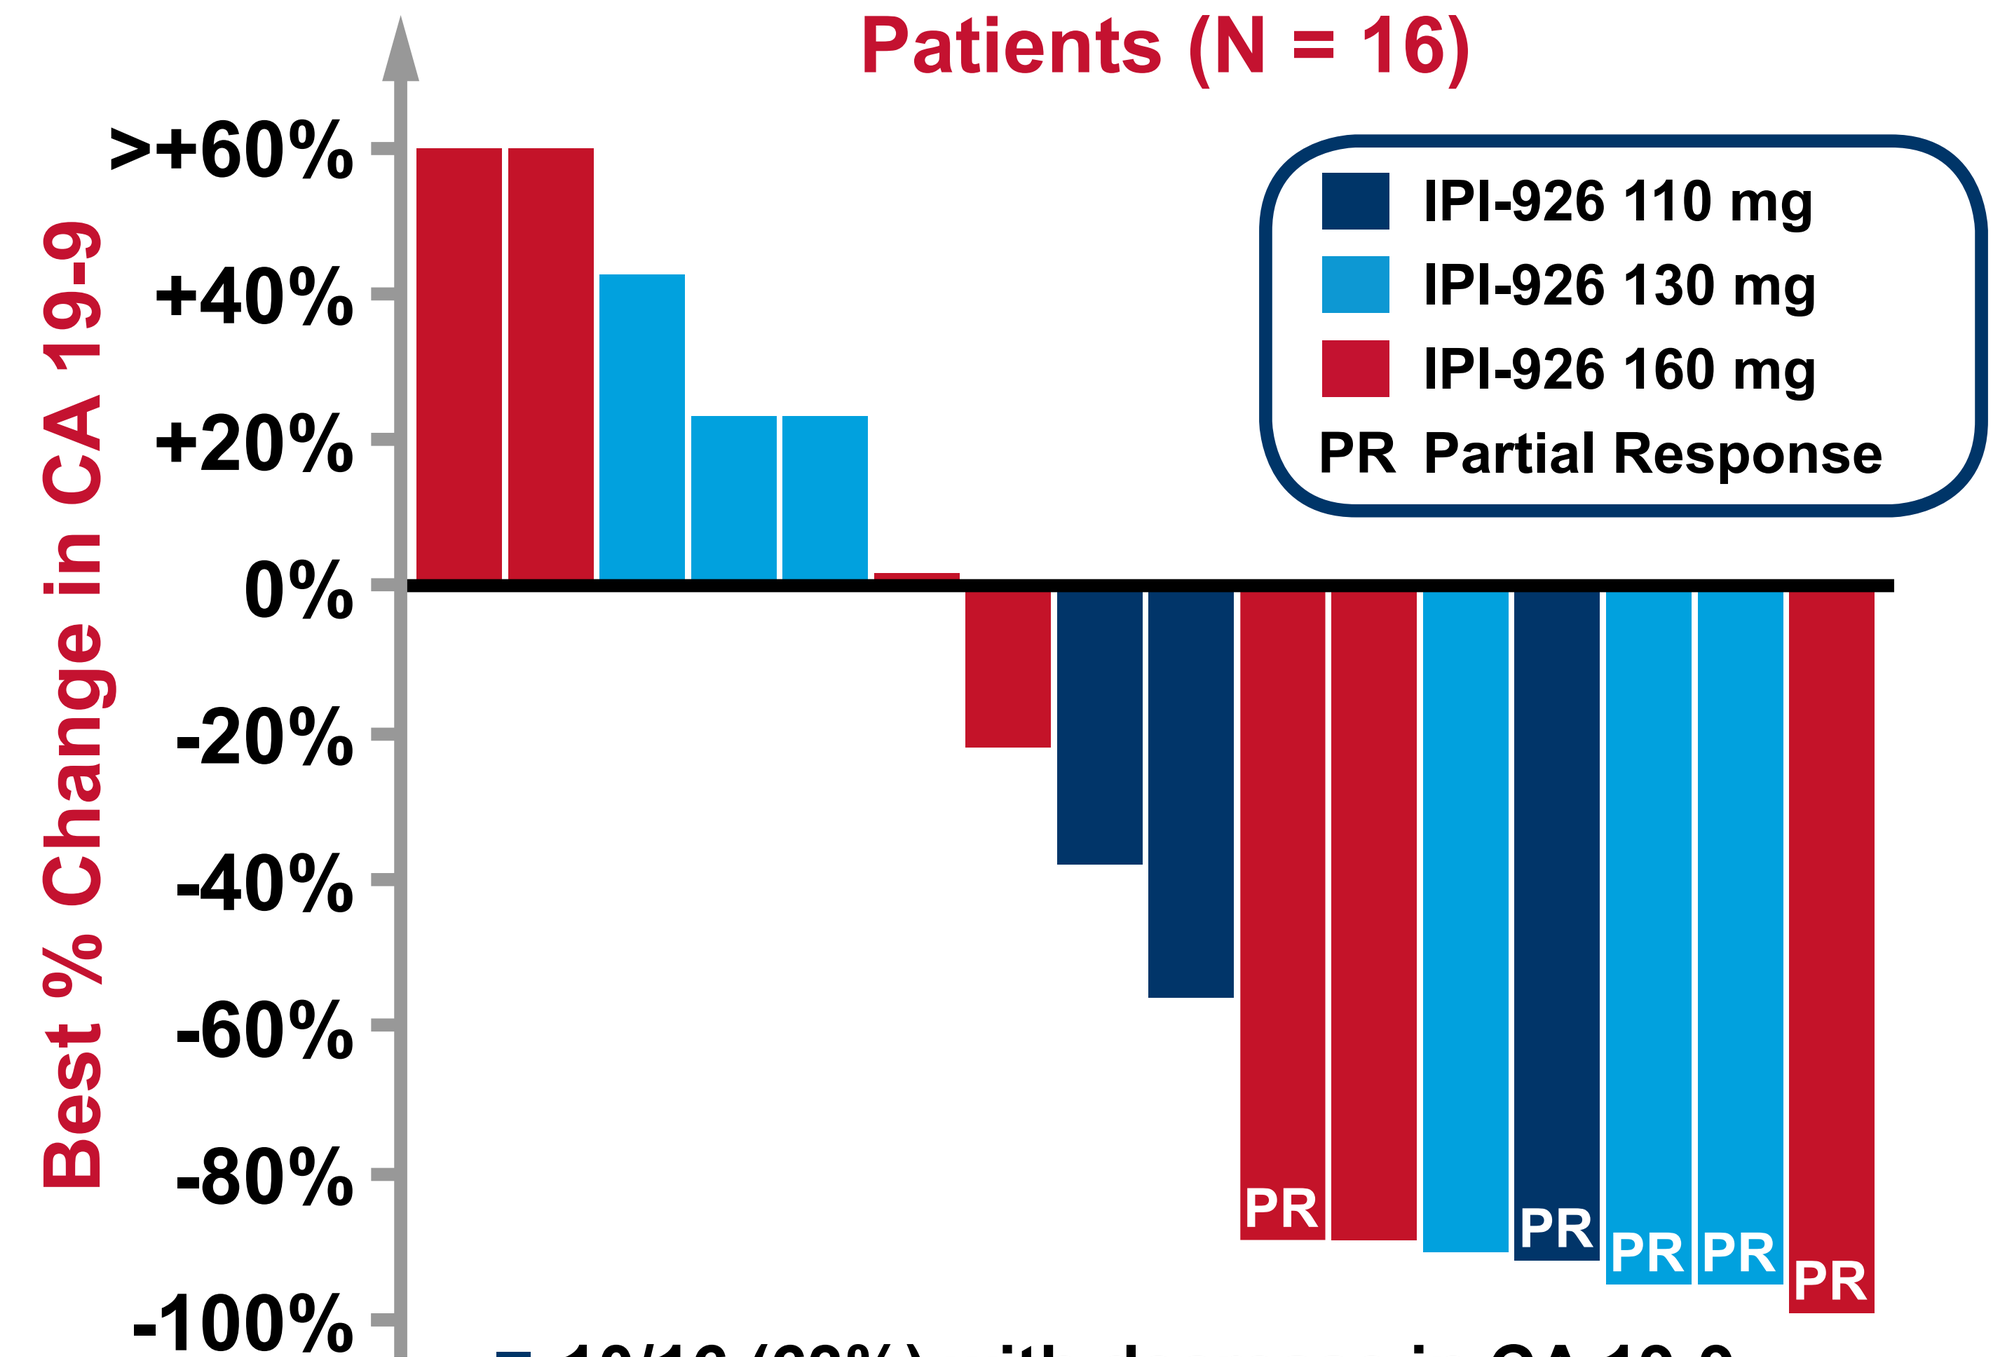
Patients (N = 16)
Best % Change in CA 19-9
>+60%
+40%
+20%
0%
-20%
-40%
-60%
-80%
-100%
IPI-926 110 mg
IPI-926 130 mg
IPI-926 160 mg
PR Partial Response
PR
PR
PR
PR
PR

- 10/16 (63%) with decrease in CA 19-9
 - 8/16 (50%) with ≥ 50% decrease
 - 5/16 (31%) with ≥ 90% decrease

Pharmacokinetics

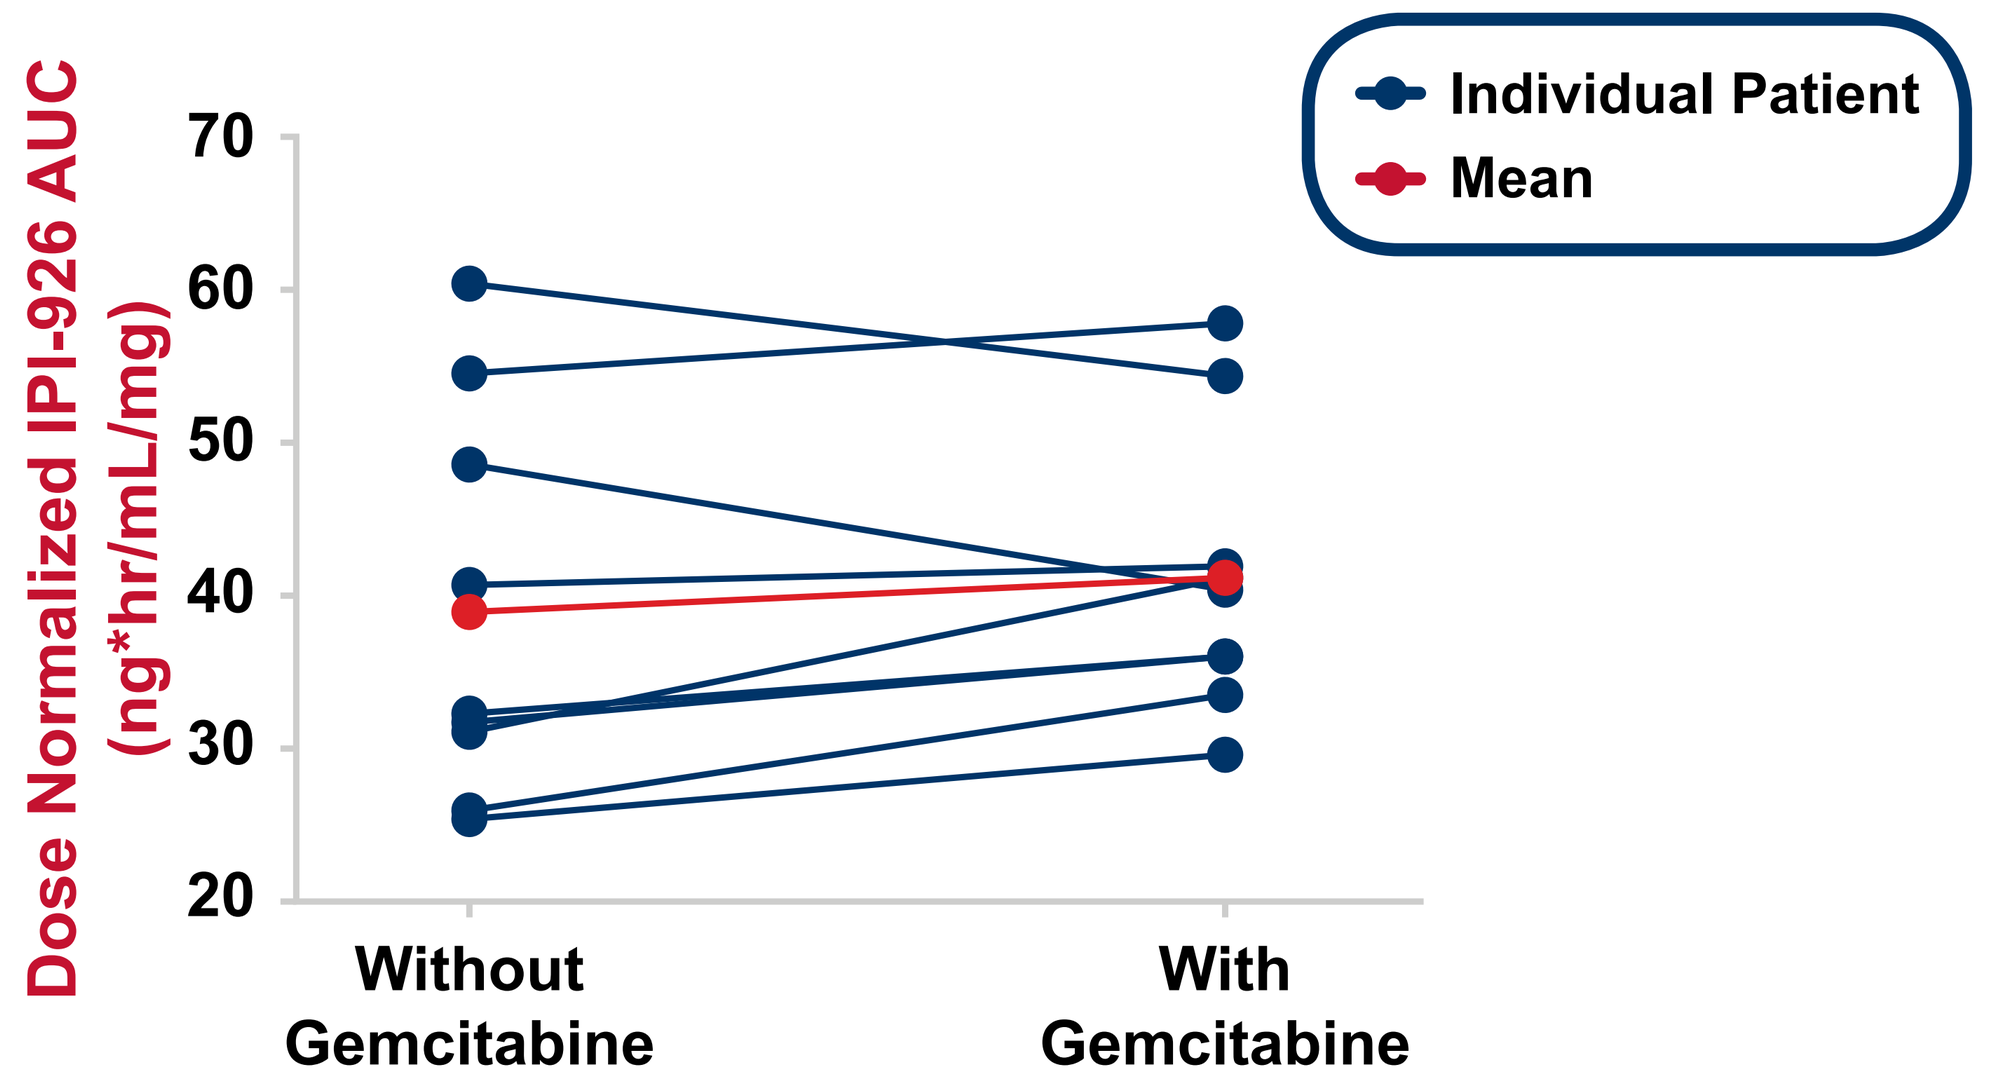
Dose Normalized IPI-926 AUC (ng*hr/mL/mg)
70
60
50
40
30
20
Without Gemcitabine
With Gemcitabine
Individual Patient
Mean

- Steady-state IPI-926 systemic exposure (AUC) is similar with and without concomitant administration of gemcitabine.

Stromal Pathway Activation

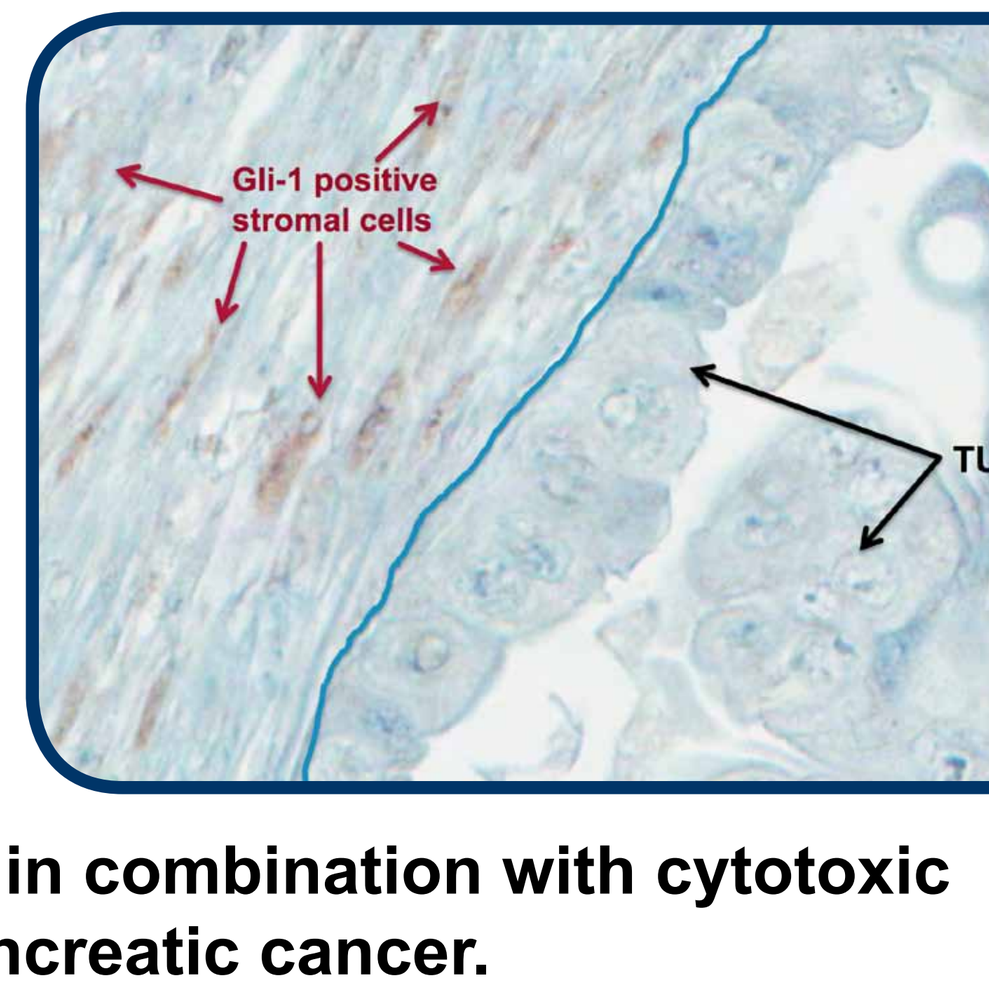
Gli-1 positive stromal cells
TUMOR

- Human pancreatic tumor tissue stained for Gli-1 (brown) demonstrates nuclear reactivity in the desmoplastic stromal cells only, consistent with active paracrine signaling in the tumor stroma.
- These data provide a compelling rationale for the investigation of IPI-926 in combination with cytotoxic therapy in patients with pancreatic cancer.

Time on Study

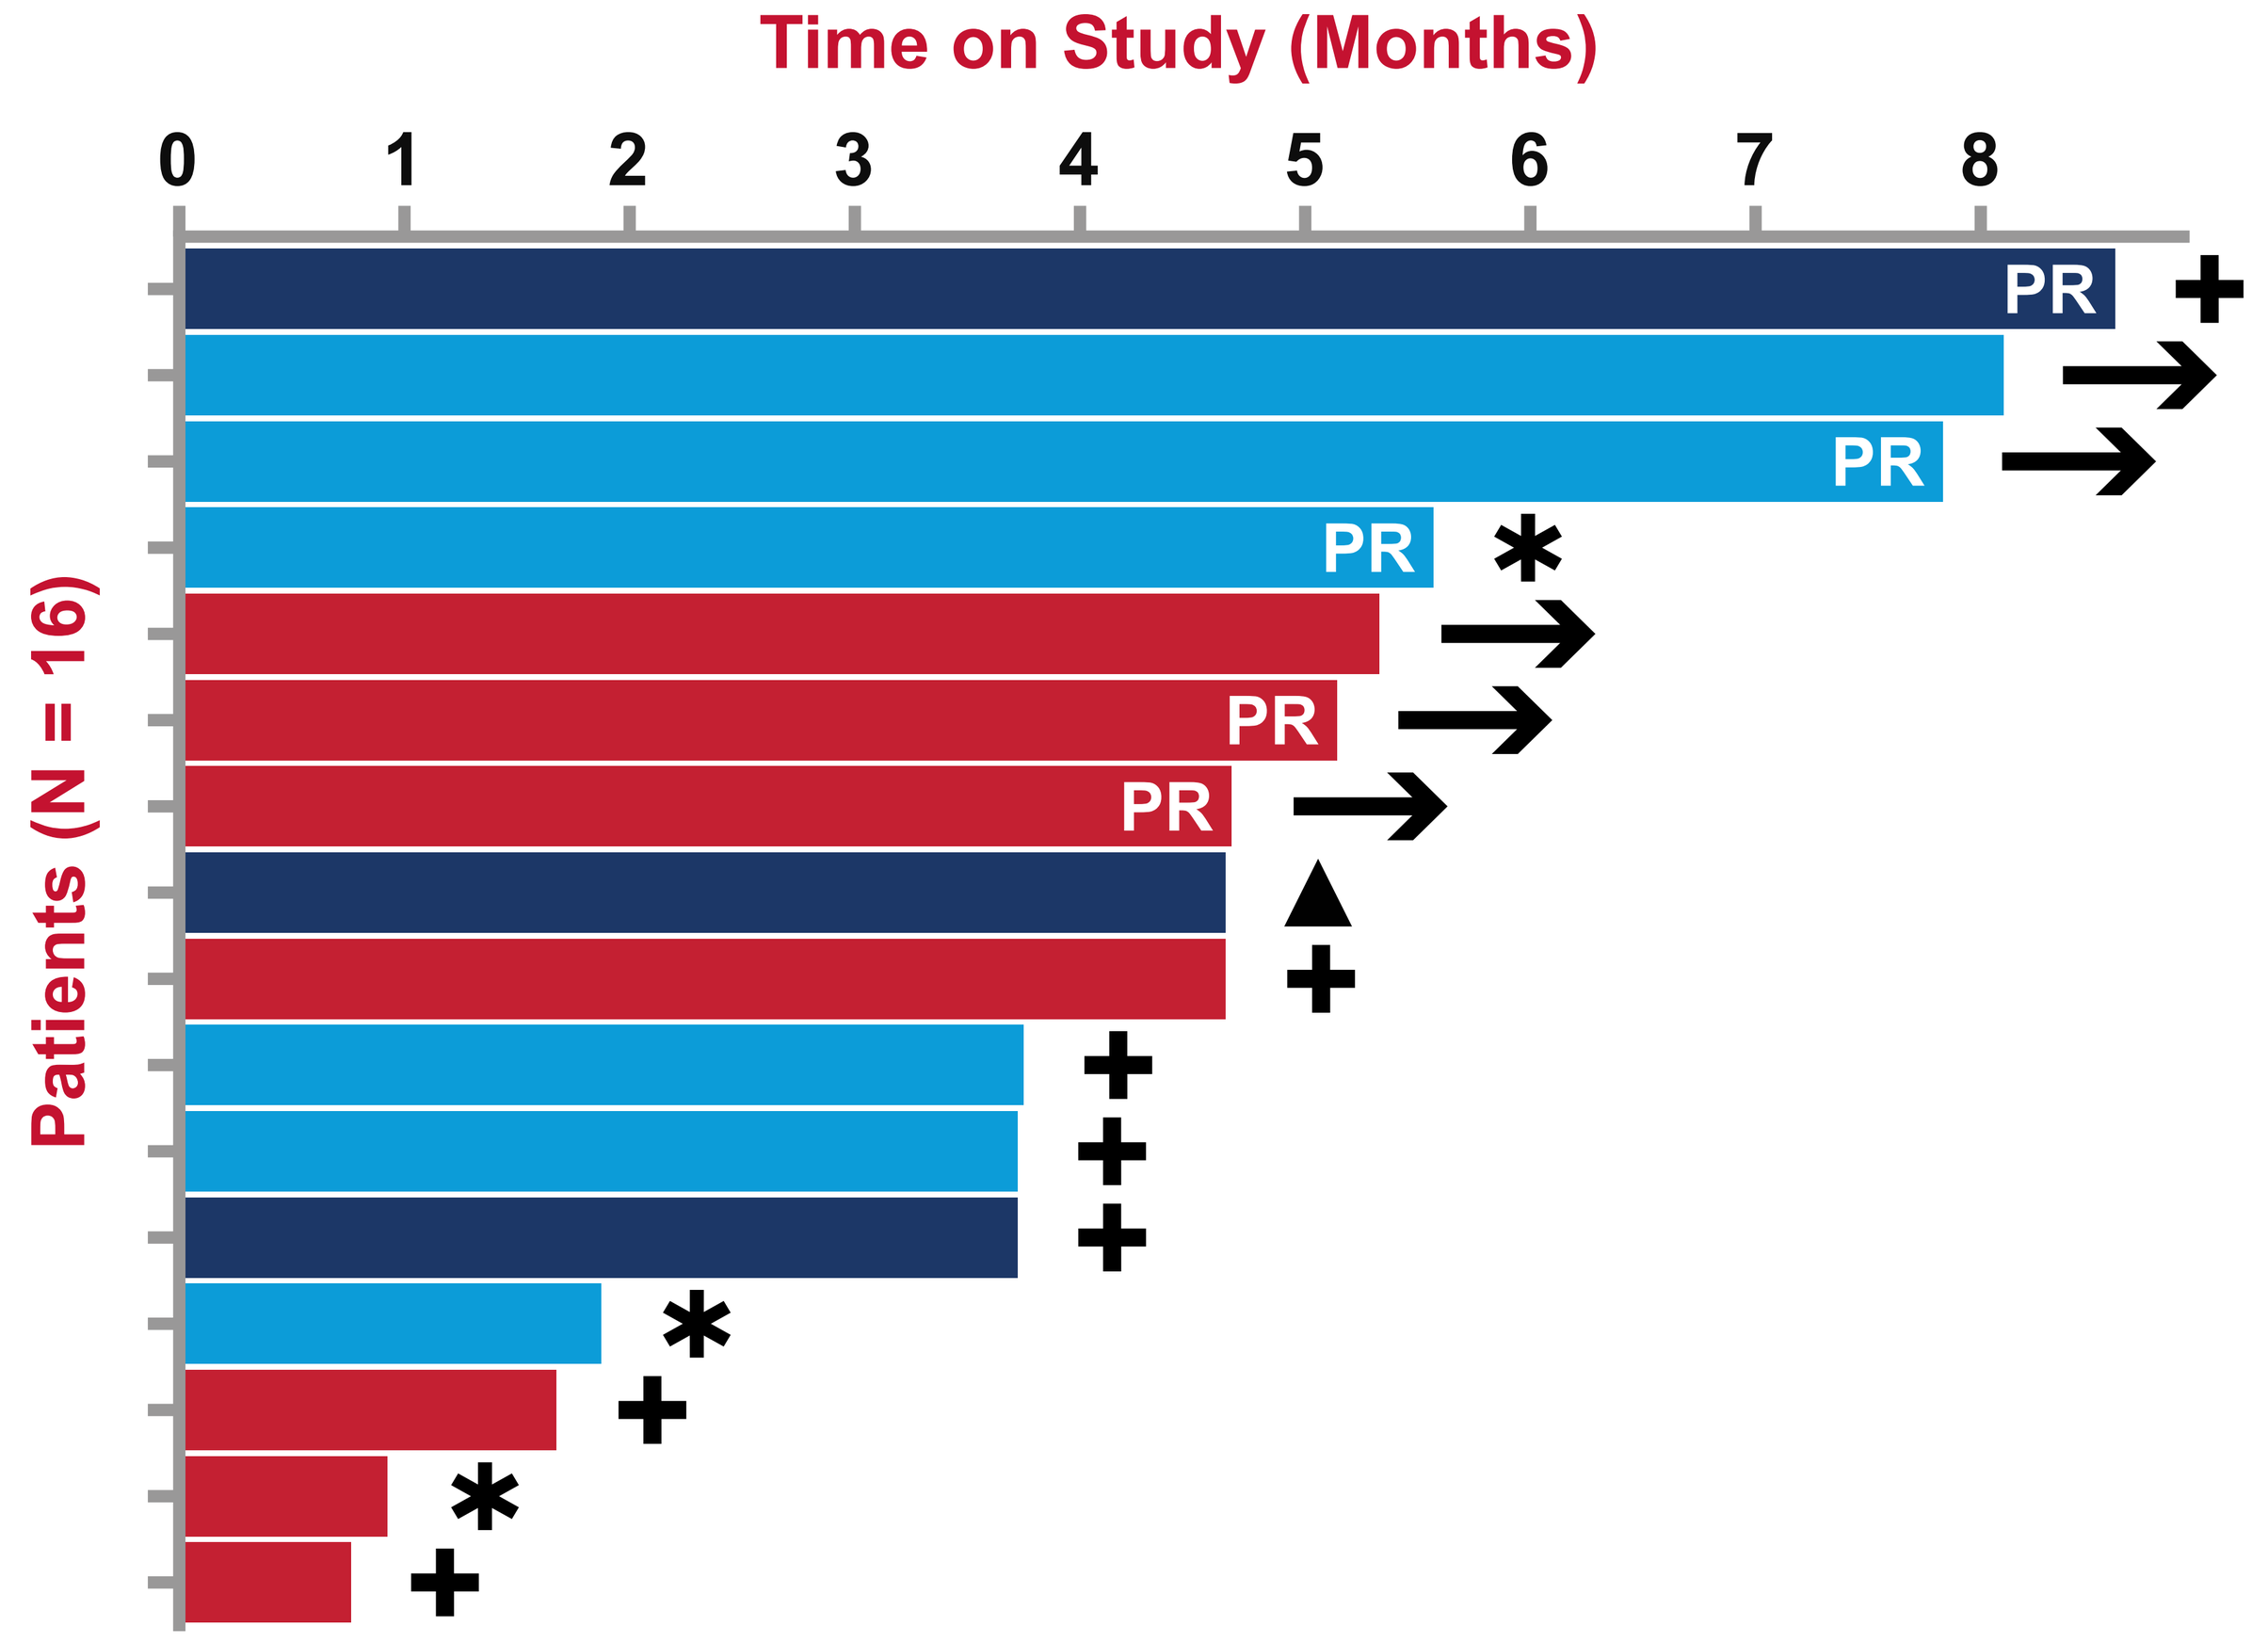
Time on Study (Months)
0
1
2
3
4
5
6
7
8
Patients (N = 16)
PR
PR
PR
PR
PR

- Median progression-free survival has not yet been reached and is ≥ 5.5 months.

IPI-926 110 mg
IPI-926 130 mg
IPI-926 160 mg
→ Patient Remaining on Study
PR Partial Response

Study Discontinuation Due To:
✚ Radiographic or Clinical Progression
▲ Adverse Event
✱ Other

Example of a Partial Response

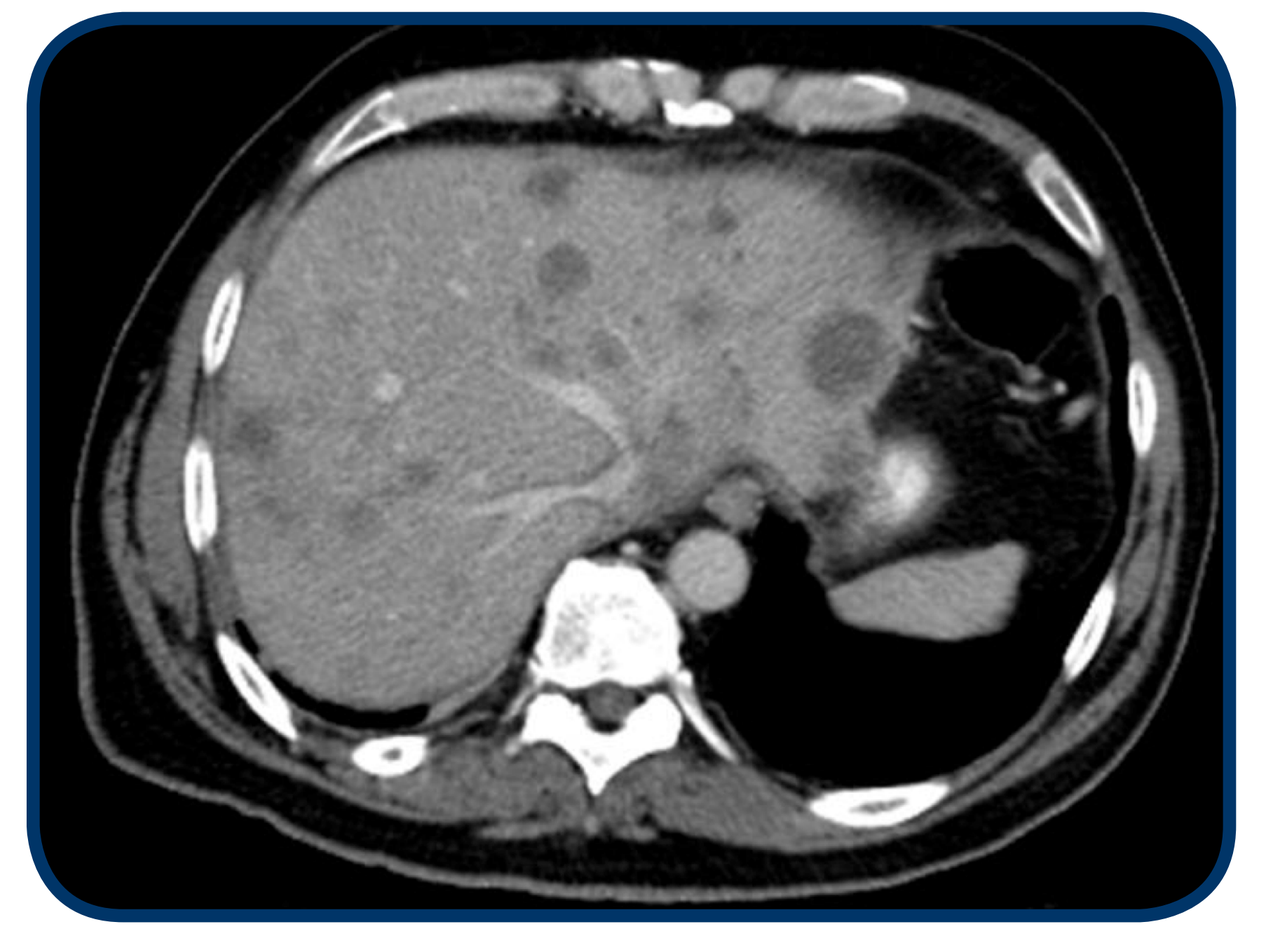

Abdominal CT:
Pre-Treatment

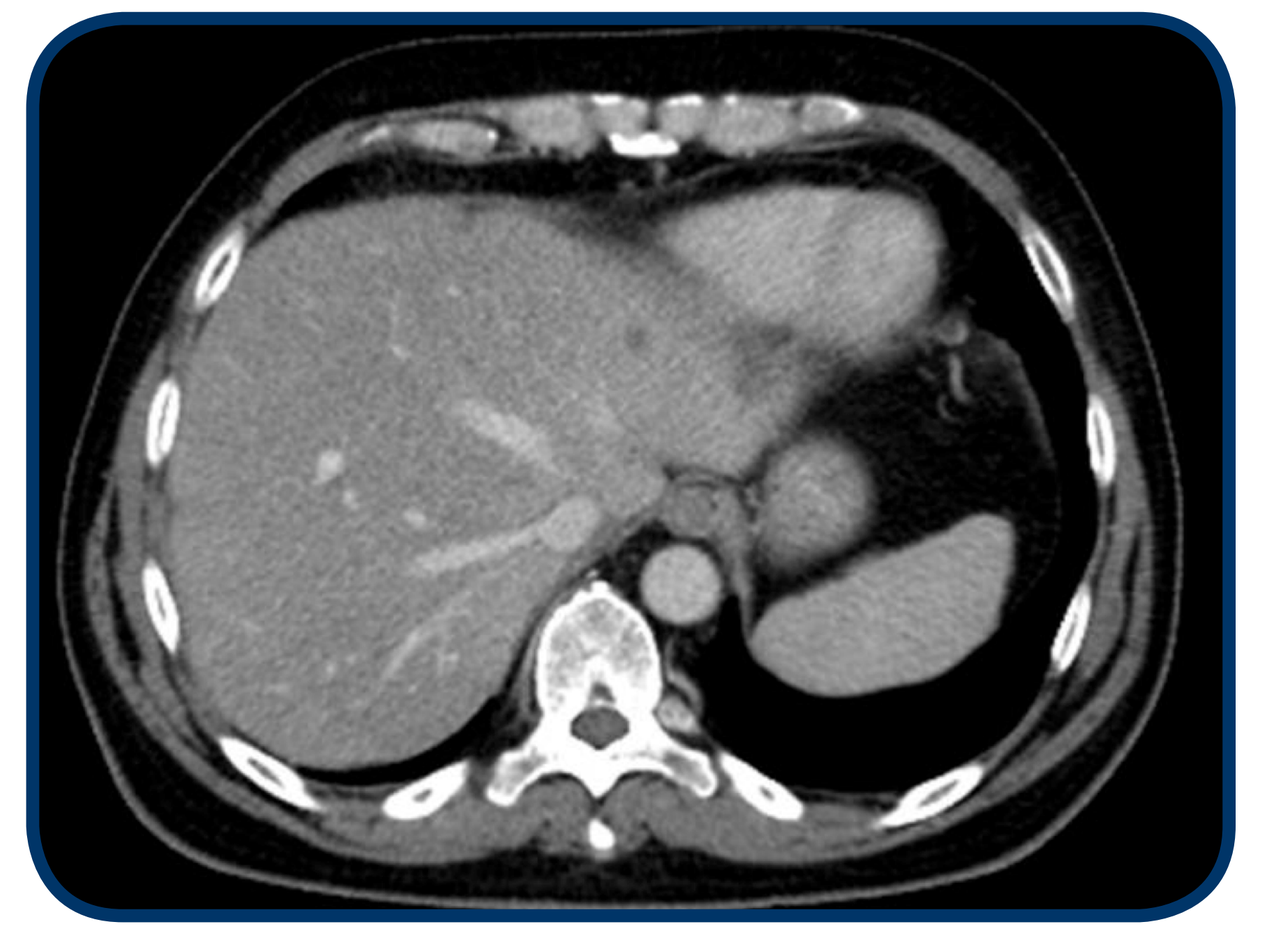

Abdominal CT:
4 Months of Treatment

- CT scan of a responding patient with metastatic pancreatic cancer after treatment with 160 mg IPI-926 and gemcitabine

Target Lesion Change

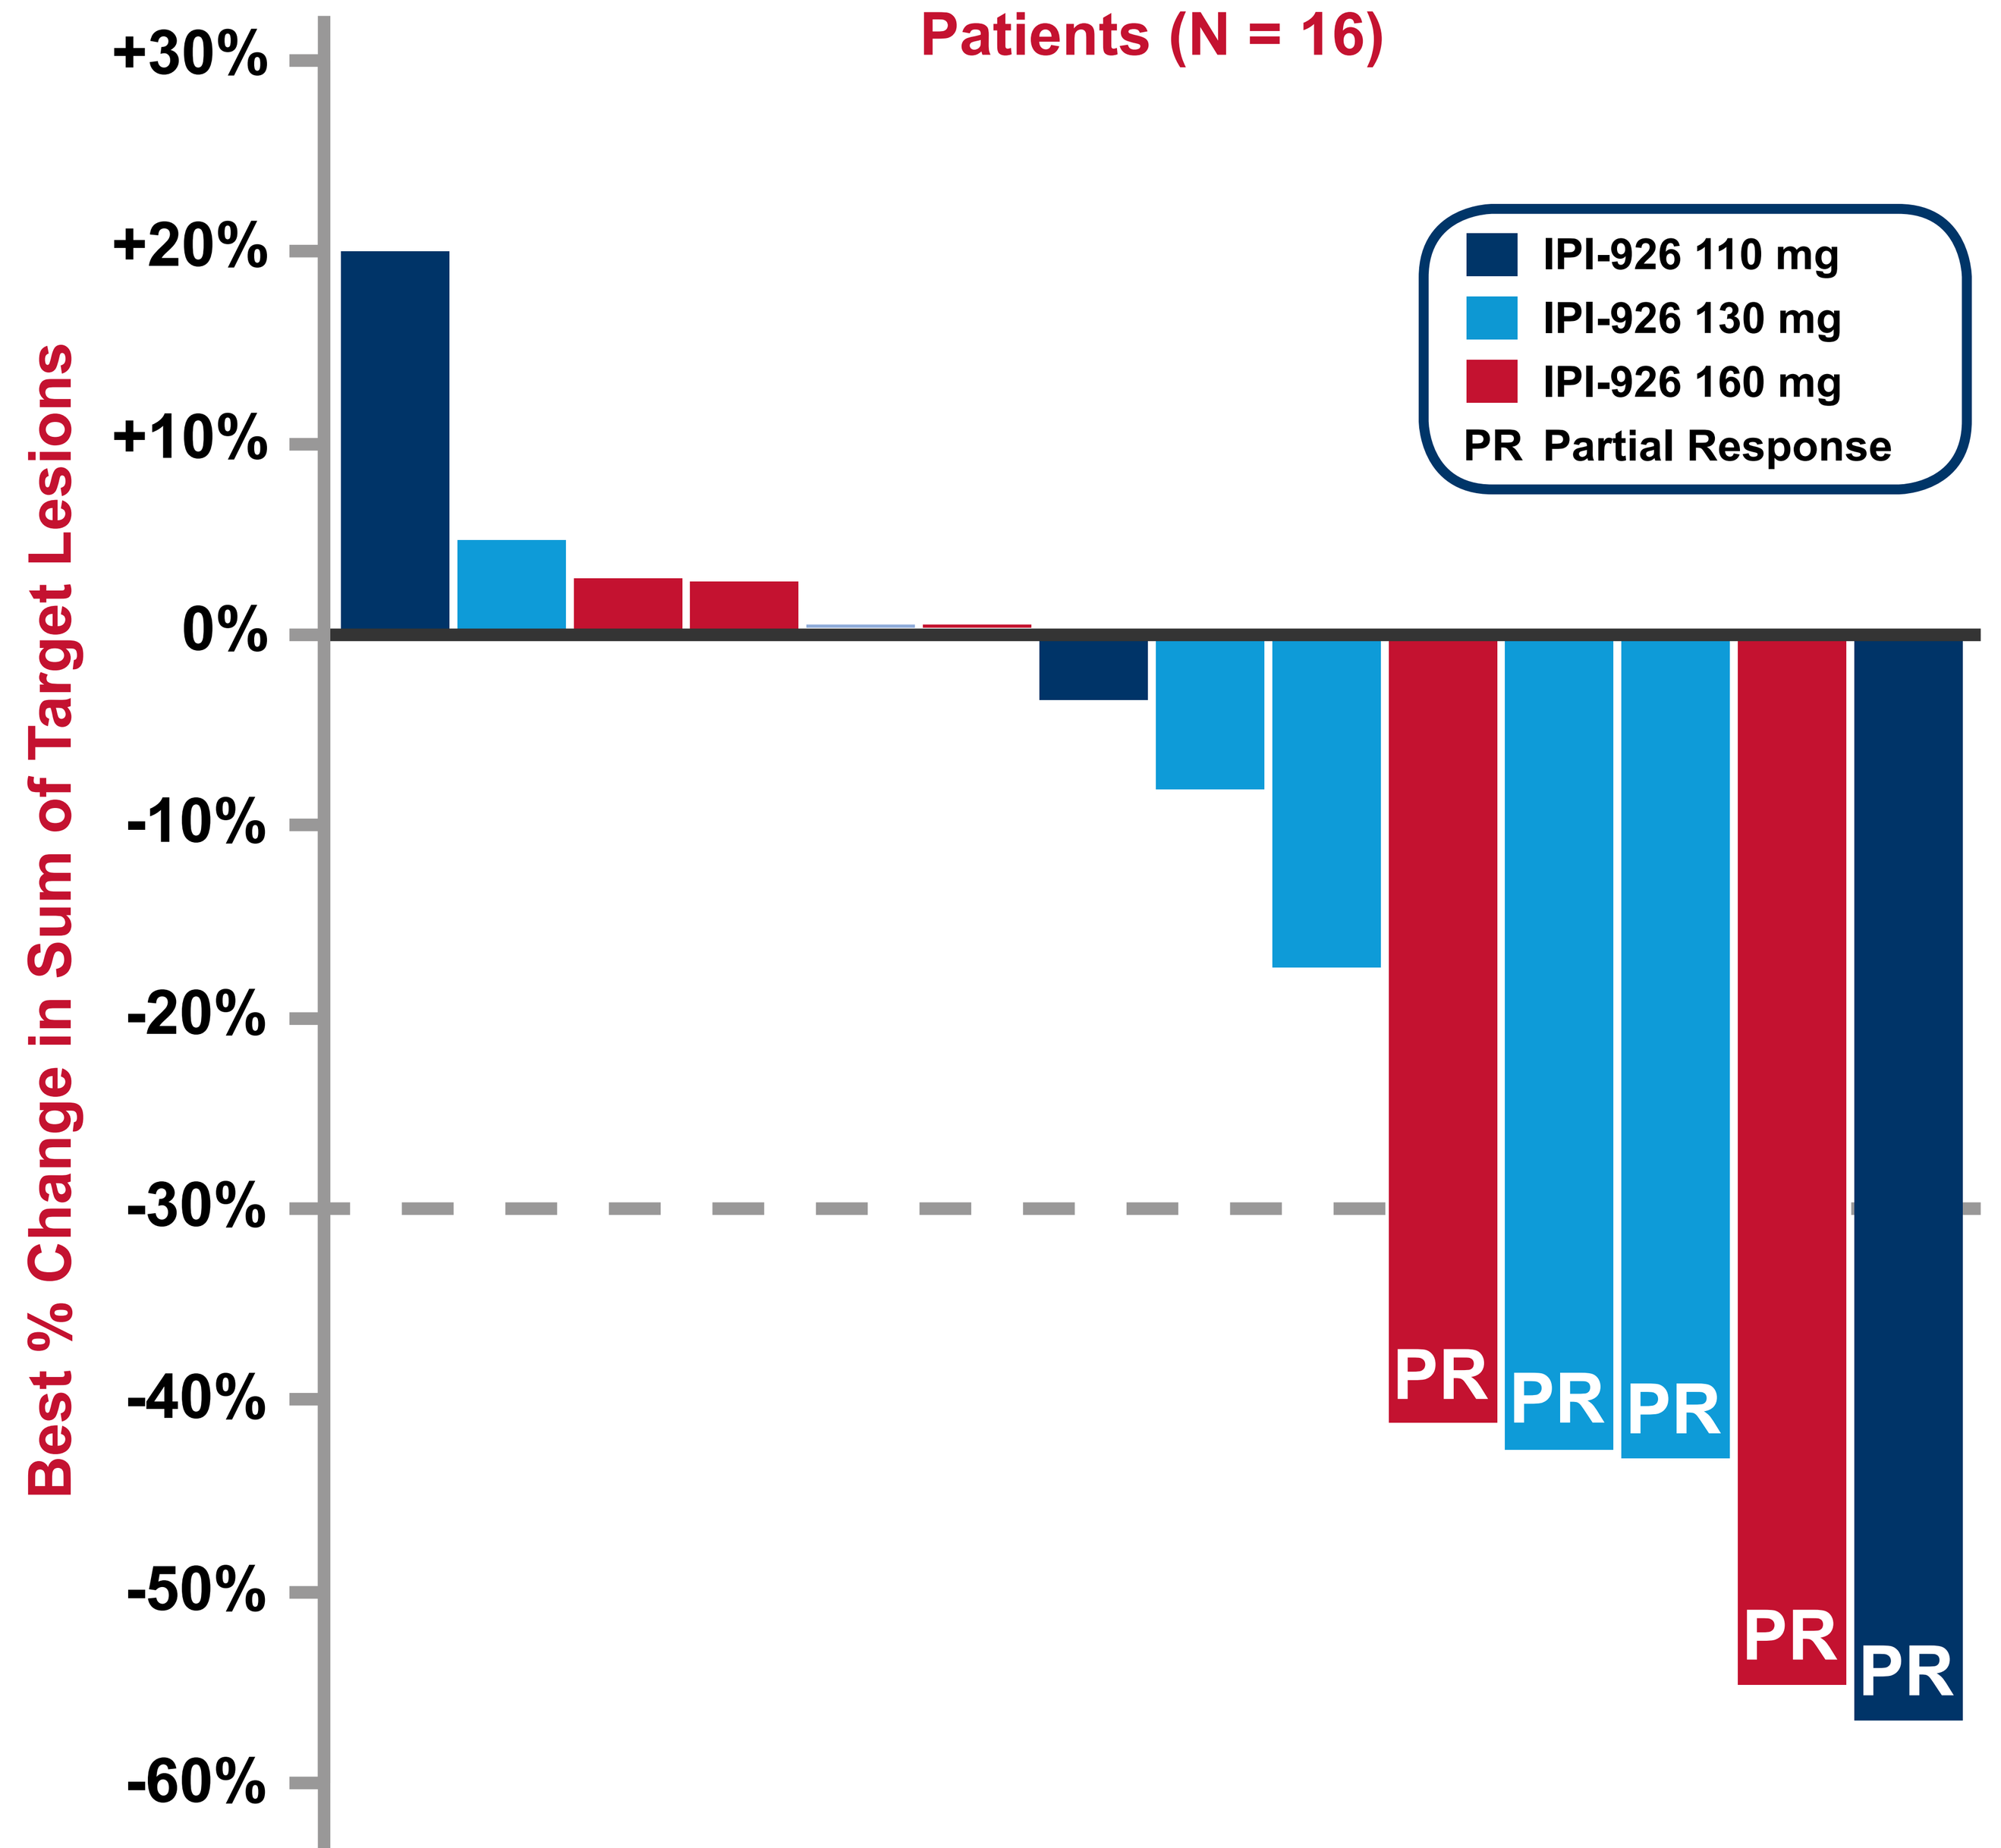
Patients (N = 16)
Best % Change in Sum of Target Lesions
+30%
+20%
+10%
0%
-10%
-20%
-30%
-40%
-50%
-60%
IPI-926 110 mg
IPI-926 130 mg
IPI-926 160 mg
PR Partial Response
PR
PR
PR
PR
PR

- 5/16 (31%) patients with partial response
- Responses observed at each dose of IPI-926 administered

Conclusions

- The combination of daily, oral IPI-926 + standard doses of gemcitabine was well-tolerated.
 - AEs observed were consistent with the known safety profile of each agent.
 - Combination did not reveal unique or more severe AEs.
- PK data demonstrated no interaction between IPI-926 and gemcitabine.
- 31% of patients had a partial response.
 - Responses were observed at each dose of IPI-926 investigated.
- The Phase 2 regimen is 160 mg IPI-926 daily with 3 weekly doses of 1000 mg/m² gemcitabine as part of a 28-day cycle.
- The Phase 2 portion of this trial is actively enrolling. This Phase 2 is randomized and placebo-controlled, has the same eligibility criteria, and has a primary endpoint of overall survival.

Poster presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, June 3-7, 2011, Chicago, Illinois