A Novel Binding Assay for Native Bcl-2 on Intact Mitochondria

Abstract #3628

Bonnie Tillotson, David Mann, Erin Brophy, Janid Ali, Vito Palombella and Christian Fritz
Infinity Pharmaceuticals, Inc., Cambridge, MA 02139

Abstract

Bcl-2 protects cells from apoptosis by binding pro-apoptotic proteins such as BID and BIM via a well defined BH3 binding groove, and is up-regulated in many forms of cancer. Significant effort has been directed to the discovery of compounds which compete with this protein-protein interaction thereby neutralizing Bcl-2's anti-apoptotic activity. Most in-vitro Bcl-2 binding assays use recombinant Bcl-2 carrying a C-terminal deletion removing it's transmembrane domain. Since in vivo, most Bcl-2 protein is found inserted into the outer mitochondrial membrane (OMM), we were interested in measuring binding to the full length Bcl-2 protein in its natural membrane environment. We therefore developed a novel Bcl-2 binding assay utilizing purified mitochondria from a human cancer cell line that carries the t(14;18) translocation, expressing large amounts of Bcl-2. The methodology measures relative binding affinity to native Bcl-2 by means of competing for a recombinant BAD-His fusion (BAD) protein. We demonstrate specific BAD binding to mitochondria purified from cells expressing Bcl-2 relative to non-Bcl-2 expressing cells. BAD binds to Bcl-2 on mitochondria in a dose dependent manner, yielding a binding isotherm with an EC50 of 20nM and a Hill coefficient close to 1. A fixed concentration of BAD protein (80nM) can be competitively displaced by BAD wt peptide with an IC50 of 20nM but not by an inactive point mutant of the BAD peptide. BIM wt peptide yields an IC50 of 1.7nM. We compared binding affinities and rank order of several published Bcl-2 binders between our new assay and assays using recombinant, truncated Bcl-2. This assay can now be used to monitor binding of putative Bcl-2 antagonists to native Bcl-2 on the OMM, providing an important link between biochemical measurements with recombinant proteins and cellular activity of Bcl-2 binding molecules.

Introduction

Bcl-2 is an important target for cancer therapy and is over-expressed in many types of cancer. It is known that Bcl-2 exhibits an anti-apoptotic effect by preventing cytochrome C release from the mitochondria and, although the precise mechanism of action of Bcl-2 is still under debate (Chen et al., 2005 and Letai et al., 2002), it is clear that the anti-apoptotic effect is mediated by protein-protein interactions involving BH3 domains. These interactions take place with Bcl-2 inserted into the outer mitochondrial membrane. Typically, determinations of ligand affinities for Bcl-2 are performed by fluorescence polarization or BIAcore assays employing truncated recombinant Bcl-2 proteins which lack the membrane insertion domain. In an effort to understand membrane associated native Bcl-2 binding and correlate this activity with the truncated form we developed a mitochondrial Bcl-2 binding assay in which exogenously added BAD protein is used as a competitive ligand to monitor Bcl-2 binding on isolated mitochondria.

Mitochondrial Bcl-2 Binding Assay: Competitive Displacement by BH3 Groove Inhibitors

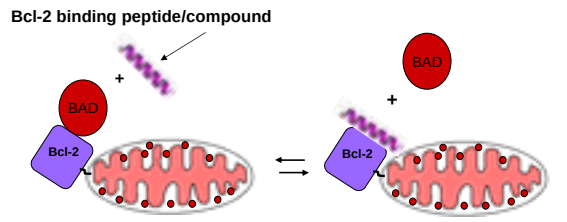

Bcl-2 binding peptide/compound
BAD
Bcl-2
BAD
Bcl-2

Materials and Methods

Mitochondrial Bcl-2 Binding Assay Schematic

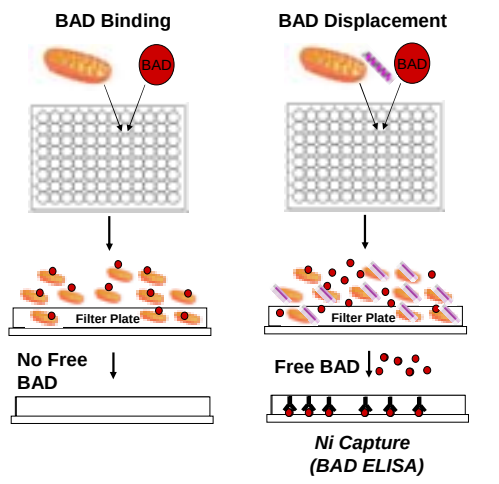

BAD Binding
BAD Displacement
BAD
BAD
Filter Plate
Filter Plate
No Free BAD
Free BAD
Ni Capture (BAD ELISA)

Step 1. Add mitochondria, BH3 peptide or compound and BAD protein to polypropylene plate. Incubate.

Step 2. Transfer mitochondria/protein mixture to Multiscreen filter plate and apply vacuum.

Step 3. Filtrate (unbound BAD-His) is captured on a HIS-Grab nickel plate. Detect BAD with standard ELISA.

Cells, reagents and antibodies:

- The human tumor cell lines RL (human follicular lymphoma) and Daudi (human Burkitt's lymphoma) were obtained from ATCC.
- Recombinant His-BAD protein (BAD) was purchased from Upstate. The wt BH3 BIM, wt BH3 BAD and mutant BH3 BAD peptides were purchased from Abgent. ABT-737 is published (Oltersdorf et al., 2005) and other Bcl-2 inhibitors were purchased from Sigma. Mouse anti-BAD and anti-BID antibodies were from BD and mouse anti-Bcl-2 HRP was from Santa Cruz. Hamster anti-mouse secondary HRP conjugate was from GE Healthcare.

Isolation of mitochondria:

- Mitochondria were isolated from cells according to a standard protocol (Letai, et al., 2002) Mitochondria were resuspended in binding buffer containing 45mM sucrose, 7.5mM TES pH 7.2, 43mM Hepes pH 7.45, 85mM KCl, 1.7mM MgCl2, 2.1mM K2HPO4, 0.2mM EGTA. The protein concentration was determined by BCA (Pierce).

BAD binding to Bcl-2 on isolated mitochondria:

- Mitochondria (15ug total protein) were preincubated with serially diluted Bcl-2 inhibitors for 15 minutes at RT. Recombinant BAD protein was added to a final concentration of 80nM and the reaction mixture incubated on ice for 1 hr, followed by filtration through a Millipore Multiscreen-HV PVDF hydrophilic filter plate to separate free BAD from mitochondria. BAD protein was quantified by an anti-His ELISA (Pierce). Mitochondrial integrity post assay was verified by cytochrome C ELISA (none detected).
- Mitochondrial Bcl-2 levels were determined by western blotting via an anti-Bcl-2 HRP antibody (Santa Cruz).

Dose Response Curve Generation

Max signal - 80 nM BAD
Min - 80 nM BAD + Mitochondria
Bound - 80 nM BAD + Mitochondria + Inhibitor

$$Inhibition = \left(\frac{Bound - Min}{Max - Min}\right)$$

IC_{50} Determination

$$Signal = \frac{A_{MAX}}{\left(\frac{IC_{50}}{I}\right)^{n} + 1}$$

Where:
Amax - Amplitude of the dose response curve
n - The Hill coefficient

Results

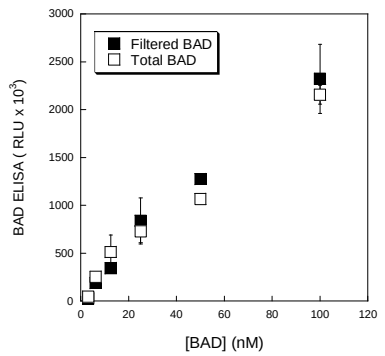

Filtered BAD
Total BAD
BAD ELISA (RLU x 10³)
[BAD] (nM)

Figure 1. BAD protein can be quantified accurately using a His-ELISA plate. Increasing concentrations of His-BAD protein were either filtered or applied directly to a nickel coated His-Grab™ plate.

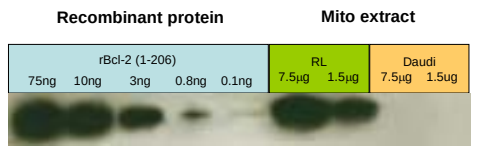

Recombinant protein
Mito extract
rBcl-2 (1-206)
75ng 10ng 3ng 0.8ng 0.1ng
RL
7.5μg 1.5μg
Daudi
7.5μg 1.5ug

Figure 2. Bcl-2 is expressed on RL but not on Daudi mitochondria. Western blot comparing RL and Daudi mitochondrial extract. In RL cells, Bcl-2 is expressed at 5μg per mg of total mitochondrial extract. No Bcl-2 expression was observed from Daudi. Neither RL nor Daudi mitochondria demonstrated detectable Bcl-xL or Bcl-w expression (data not shown).

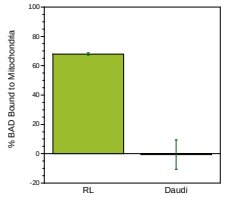

% BAD Bound to Mitochondria
RL
Daudi

Figure 3. BAD protein binds specifically to mitochondria expressing Bcl-2. 70% of the added His-BAD protein bound to RL mitochondria whereas no binding was observed to Daudi mitochondria.

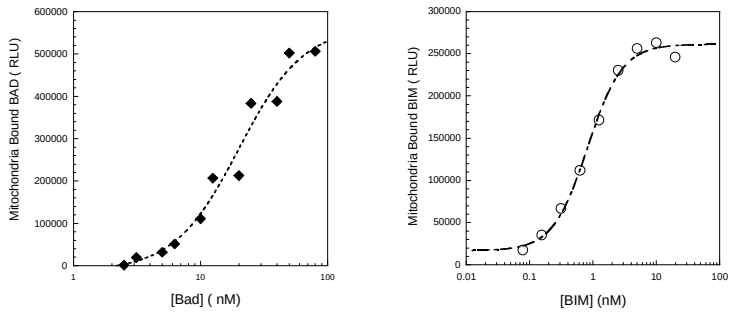

Mitochondria Bound BAD (RLU)
[Bad] (nM)
Mitochondria Bound BIM (RLU)
[BIM] (nM)

BH3 protein	EC_{50} (mitochondrial binding)	Ki (fluorescence polarization)
BAD	20 +/- 4.2nM	11.2 +/- 2.4nM
BIM	0.8 +/- 0.08nM	0.1 +/- 0.2nM

Figure 4. BIM protein has a higher affinity for Bcl-2 on mitochondria than BAD. The higher affinity of BIM to endogenous Bcl-2 on RL mitochondria correlates with the affinity trend observed for recombinant Bcl-2 in an FP binding assay.

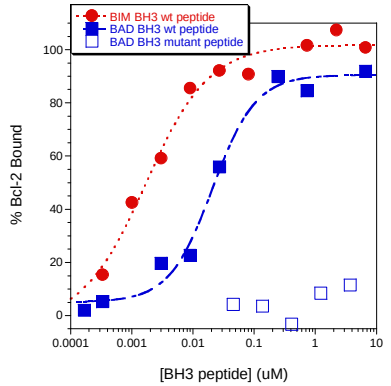

BIM BH3 wt peptide
BAD BH3 wt peptide
BAD BH3 mutant peptide
% Bcl-2 Bound
[BH3 peptide] (uM)

Figure 5. BAD displacement assay: Specific binding of BH3 peptides to Bcl-2 on mitochondria. BAD displacement can be used to measure specific BH3 peptide binding to Bcl-2 on RL mitochondria. WT BAD and WT BIM peptide bind to mitochondria by displacing BAD with an IC_{50} of 20 (+/- 10)nM and IC_{50} of 1.7 (+/- 2)nM respectively. A mutant BAD BH3 peptide was inactive (>10μM). Averages reported are from triplicate data.

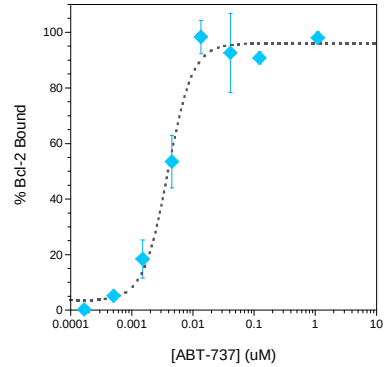

% Bcl-2 Bound
[ABT-737] (uM)

Figure 6. ABT-737 binds potently to Bcl-2 on Mitochondria. ABT-737 binds to Bcl-2 on RL mitochondria as measured by BAD protein displacement with an IC_{50} of 2.4 (+/-0.6)nM. Averages reported are from duplicate data.

Bcl-2 Inhibitors Assayed

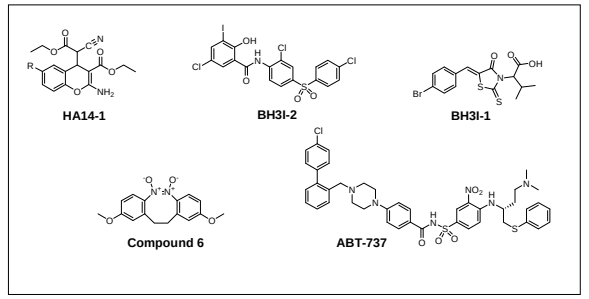

HA14-1
BH3I-2
BH3I-1
Compound 6
ABT-737

Figure 7. Published small molecules reported to bind to Bcl-2. (*Reed and Pellecchia, 2005*)

Bcl-2 Binder	Mitochondrial Binding IC_{50} (μM)	Fluorescence Polarization Ki (μM)
ABT-737	0.0024	< 0.001
Bim BH3 WT peptide	0.0017	< 0.001
BAD BH3 WT peptide	0.02	0.011
Compound 6	39% @ 100μM	29
BH31-2'	38% @ 100μM	24
BH31-1	37% @ 100μM	3.6
HA14-1	11% @ 100μM	50
BAD BH3 mutant peptide	>100μM	>30μM

Table 1. Comparison of mitochondrial Bcl-2 binding vs. binding to recombinant protein measured by FP. Affinities of known Bcl-2 binders were compared in the mitochondrial Bcl-2 binding assay to FP values generated with recombinant Bcl-2 (Certo et al., 2006 and Oltersdorf et al., 2005). A similar ranking of affinities was observed. The lower limit of detection (IC50 values) for the mitochondrial Bcl-2 binding assay is 1-2nM. Data is representative of duplicate measurements.

Summary

- **Mitochondria from RL cells express Bcl-2 whereas mitochondria from Daudi cells do not**
- **Bcl-2 expressed on RL mitochondria binds specifically to BH3 only proteins**
- **Displacement of BAD protein can be used to measure binding to Bcl-2 on mitochondria**
- **Using this system, we are able to measure binding of peptides and small molecules to Bcl-2 on mitochondria**
- **These results show that binding to endogenous Bcl-2 inserted into the mitochondrial membrane correlate well with data obtained using truncated, recombinant Bcl-2**
- **This method allows assessment of ligand binding to Bcl-2 in a more physiologically relevant environment**

References

- Michael Certo, Victoria Del Gaizo Moore, Mari Nishino, Guo Wei, Stanley Korsmeyer, Scott A. Armstrong and Anthony Letai, *Mitochondria primed by death signals determine cellular addiction to antiapoptotic BCL-2 family members*. Cancer Cell (2006) 9, 351-365
- Lin Chen, Simon N. Willis, Andrew Wei, Brian J. Smith, Jamie I. Fletcher, Mark G. Hinds, Peter M. Colman, Catherine L. Day, Jerry M. Adams, and David C.S. Huang. *Differential Targeting of Prosurvival Bcl-2 proteins by their BH3-Only ligands allows complementary apoptotic function.* Molecular Cell (2005) 17, 393–403
- Anthony Letai, Michael C. Bassik, Loren D. Walensky, Mia D. Sorcinelli, Solly Weiler and Stanley J. Korsmeyer, *Distinct BH3 domains either sensitize or activate mitochondrial apoptosis, serving as prototype cancer therapeutics.* Cancer Cell (2002) 2, 183-192
- Tilman Oltersdorf, Steven W. Elmore, Alexander R. Shoemaker, Robert C. Armstrong, David J. Augeri, Barbara A. Belli, Milan Bruncko, Thomas L. Deckwerth, Jurgen Dinges, Philip J. Hajduk, Mary K. Joseph, Shinichi Kitada, Stanley J. Korsmeyer, Aaron R. Kunzer, Anthony Letai, Chi Li, Michael J. Mitten, David G. Nettesheim, ShiChung Ng, Paul M. Nimmer, Jacqueline M. O'Connor, Anatol Oleksijew, Andrew M. Petros, John C. Reed, Wang Shen, Stephen K. Tahir, Craig B. Thompson, Kevin J. Tomaselli, Baole Wang, Michael D. Wendt, Haichao Zhang, Stephen W. Fesik and Saul H. Rosenberg, *An inhibitor of Bcl-2 family proteins induces regression of solid tumours.* Nature Letters (2005) 435, 677-681
- John C. Reed and Maurizio Pellecchia. *Apoptosis-based therapies for hematologic malignancies,* Blood (2005) 106, 408-418