Depilation Induced Anagen as a Model to Study Hedgehog Pathway Antagonist IPI-926 Implications for Biomarker Development

Abstract # 2827

Kerrie Faia, Everton Mandley, Suzy Dembski, Christine Pien, John MacDougall, Jennifer Hoyt, Matthew Campbell, Grace Lin, Priscilla Lo, Margaret Read and Karen McGovern.

Infinity Pharmaceuticals Inc., 780 Memorial Drive, Cambridge, Massachusetts, USA

ABSTRACT

The Hedgehog cell signaling pathway is normally active during embryonic development and plays a critical role in controlling the growth and differentiation of pluripotent progenitor cells in many tissues, including the skin. The Sonic Hedgehog ligand (SHH) is a key regulator of hair follicle growth and cycling and serves as a switch between the resting (telogen) and the growth (anagen) stage of the hair cycle. The hair cycle in the C57BL/6 mouse has been extensively characterized and post depilation provides a highly standardized model in which to explore Hedgehog pathway biology. Using 7 week old C57BL/6 mice which are in the telogen phase of the hair cycle, anagen and subsequently hair re-growth were initiated via chemical depilation with Nair. As a result of anagen initiation, the hair follicle cycle is synchronized allowing for reproducible measurement of Hedgehog target gene expression over time in the skin. Shaved skin, containing hair follicles which remain in telogen, serves as a control. From days 6 through 14 post depilation, corresponding to mid to late anagen, Hedgehog target gene expression was measured by RT-PCR. The Hedgehog signaling pathway is active during anagen as SHH, Gli-1, Gli-2, HHIP and PTCH-1 were all up-regulated in the depilated but not the shaved skin samples. The highest level of Hedgehog target gene expression was noted on day 10 post depilation. Smoothened (SMO) levels remained constant throughout the study and did not differ between telogen and anagen. Having established the up-regulation of Hedgehog target genes during depilation induced anagen, our novel SMO antagonist IPI-926 was evaluated in this model. IPI-926 is an orally bioavailable cyclopamine derivative with favorable PK properties and is a potent inhibitor of the Hedgehog pathway. To test compound activity, a single oral dose of either vehicle or IPI-926 was administered on day 10 post depilation. At various time points post dose, both shaved and depilated skin samples were collected to evaluate gene expression. In a dose-proportional manner, IPI-926 completely inhibited GLI-1 up-regulation in the depilated skin as early as 8 hours post dose and maintains complete inhibition out to 48 hours. IPI-926 also inhibits GLI-1 expression induced as a result of natural anagen, which occurs at approximately 12 weeks of age. Of note, after hair follicle synchronization, onset of melanogenesis occurs on Day 9 post depilation and hair re-growth by day 14. Daily administration of IPI-926 or BID administration of the SHH blocking antibody, 5E1, both inhibited hair re-growth post depilation. Collectively this data suggests that Hedgehog target gene expression and regulation in the hair follicle offers an attractive and obtainable biomarker for Hedgehog pathway antagonists under evaluation in the clinic as anti-cancer agents.

MODEL DEVELOPMENT

Depilation Induced Hair Growth as a Model to Study the Hedgehog Pathway

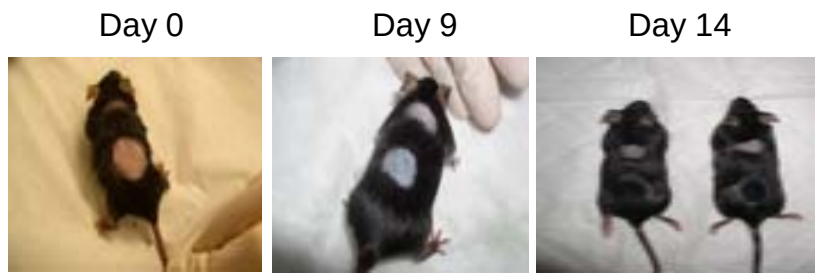

Day 0
Day 9
Day 14

Using 7 week old C57BL/6 mice that are in the telogen (resting) phase of the hair cycle, fur was removed either by shaving alone (top spot) or by depilation with Nair ™ (bottom spot). On day 9 post depilation, skin darkening, an indicator of follicular melanogeneisis, is evident. Two weeks post depilation, the fur has grown back on the depilated area only.

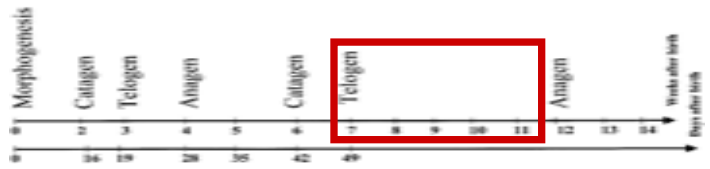

Morphogenesis
Catagen
Telogen
Anagen
Catagen
Telogen
Anagen

Muller-Rover *et al*. Journal of Investigational Dermatology 2001

Induction of Hedgehog Target Gene Gli-1 During Anagen is Maximal on Day 10 Post Depilation

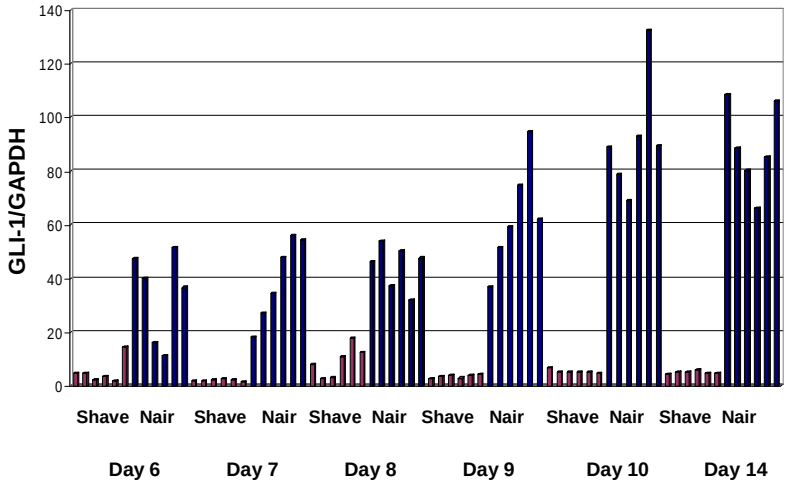

GLI-1/GAPDH
140
120
100
80
60
40
20
0
Shave Nair Shave Nair Shave Nair Shave Nair Shave Nair Shave Nair
Day 6
Day 7
Day 8
Day 9
Day 10
Day 14

To obtain a time course of hedgehog target gene expression post depilation, 36 mice were both shaved and depilated. On days 6 through 14 post depilation, 6 mice were sacrificed and their skin collected for RT-PCR analysis. Expression of Gli-1, Gli-2, HHIP and PTCH-1 were all upregulated during anagen while SMO levels remained constant throughout the study.

DISCOVERY OF IPI-926

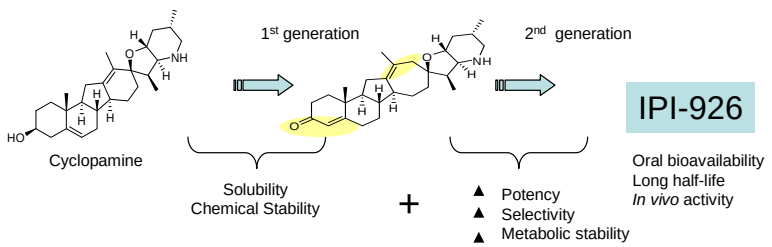

Cyclopamine
1st generation
2nd generation
IPI-926
Solubility
Chemical Stability
+
Potency
Selectivity
Metabolic stability
Oral bioavailability
Long half-life
In vivo activity

PD RESPONSE OF IPI-926

IPI-926 can maintain pathway inhibition for 48 hrs

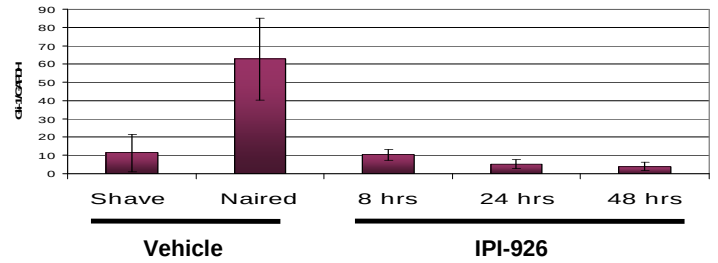

Gli-1/GAPDH
90
80
70
60
50
40
30
20
10
0
Shave
Naired
8 hrs
24 hrs
48 hrs
Vehicle
IPI-926

A single dose of either vehicle or 100mg/kg IPI-926 was administered on day 10 post depilation. Shaved and depilated skin was collected for Gli-1 expression analysis by RT-PCR. Depilation with Nair™ induced Gli-1 expression 6 fold compared to shaved skin samples. This induction was completely inhibited by IPI-926 and was maintained for the duration of the study. N=6 mice per group.

Dose Responsive Inhibition of Gli-1 with IPI-926

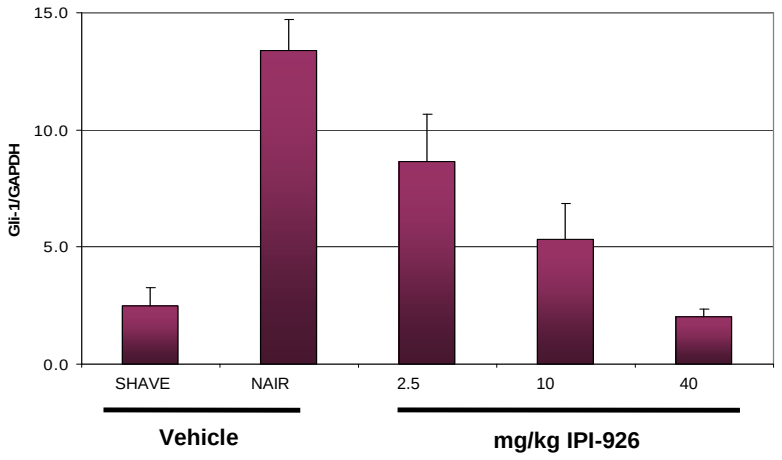

Gli-1/GAPDH
15.0
10.0
5.0
0.0
SHAVE
NAIR
2.5
10
40
Vehicle
mg/kg IPI-926

On day 10 post depilation, mice were treated with a single dose of either vehicle or IPI-926. Skin was collected 8 hours post dose for GLI-1 analysis by RT-PCR. N=6 mice per group.

IPI-926 reduces Gli-1 expression during naturally occurring anagen

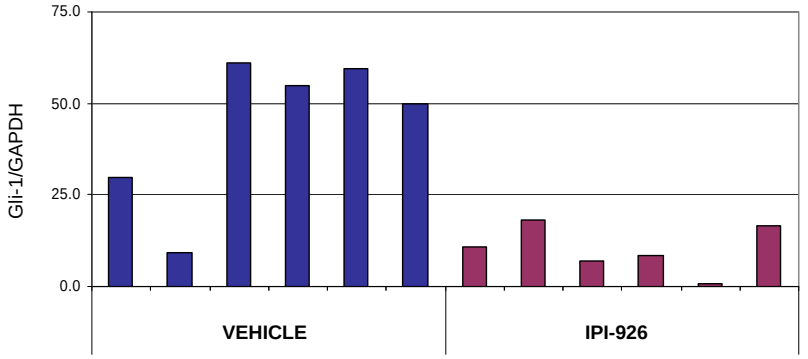

Gli-1/GAPDH
75.0
50.0
25.0
0.0
VEHICLE
IPI-926

To determine if IPI-926 would inhibit Gli-1 expression as a result of naturally occurring anagen, adult mice between 12 and 14 weeks of age were treated with a single dose of either vehicle or IPI-926 (100mg/kg). At 8 hours post dose, skin was collected for Gli-1 expression analysis by RT-PCR. Data from the individual mice are shown.

Hair Cycle in C57Bl/6 mice

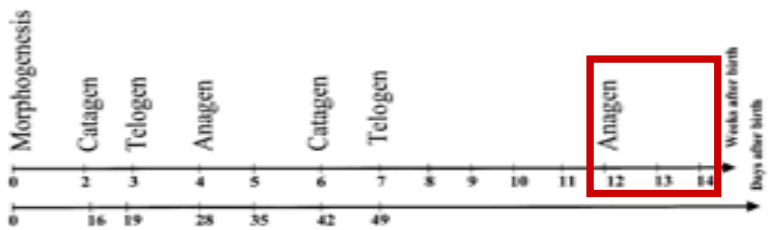

Morphogenesis
Catagen
Telogen
Anagen
Catagen
Telogen
Anagen

BIOLOGICAL ACTIVITY OF IPI-926 AND 5E1 ARE SIMILAR

Both IPI-926 and 5E1 inhibit Gli-1 induction And hair re-growth post depilation

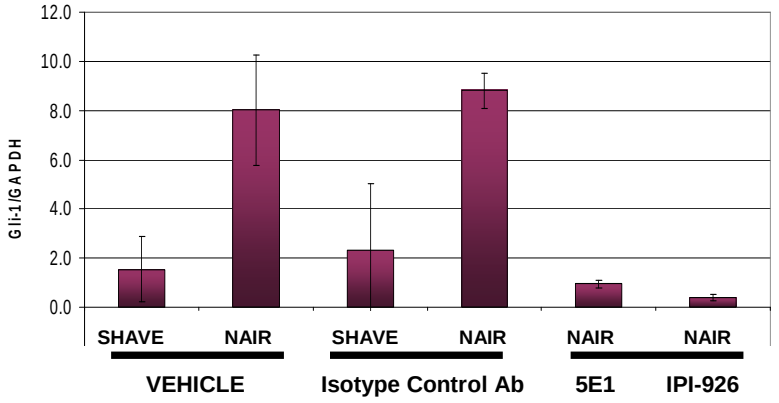

Gli-1/GAPDH
12.0
10.0
8.0
6.0
4.0
2.0
0.0
SHAVE
NAIR
SHAVE
NAIR
NAIR
NAIR
VEHICLE
Isotype Control Ab
5E1
IPI-926

To determine if inhibition of the Hedgehog pathway would prevent hair re-growth post depilation, 36 mice were shaved and depilated at 7 weeks of age. Mice received daily doses of either Vehicle or IPI-926 (40mg/kg), or were dosed every other day with the 1A7 isotype control or the SHH blocking monoclonal antibody 5E1 at 6mg/kg. Drug administration commenced on the day of depilation. At the end of the study (day 16 post depilation) skin was collected for RT-PCR.

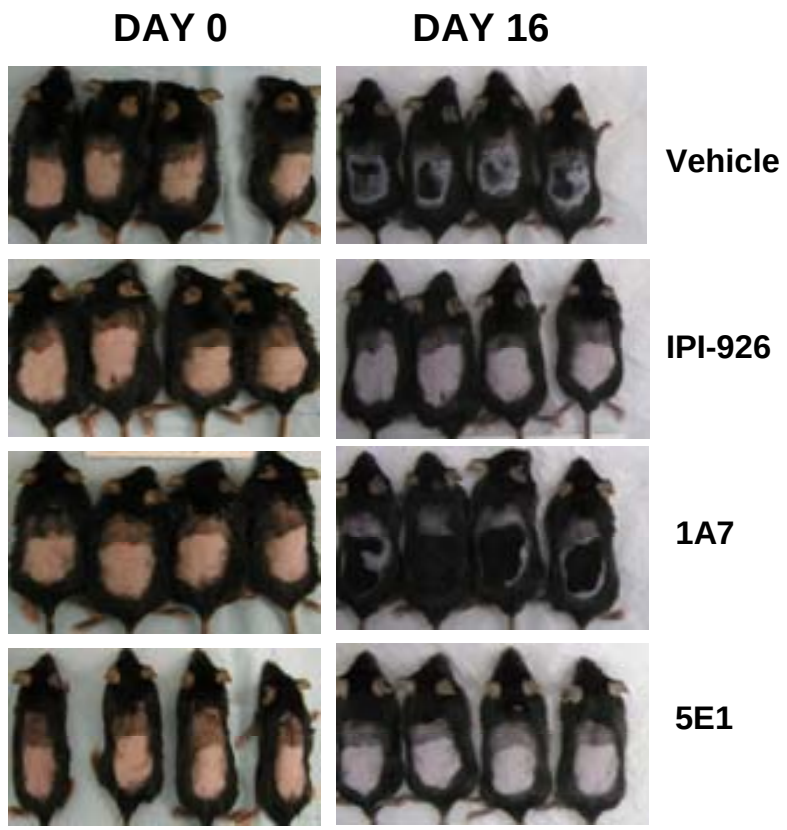

DAY 0
DAY 16
Vehicle
IPI-926
1A7
5E1

HEDGEHOG GENE SIGNATURE

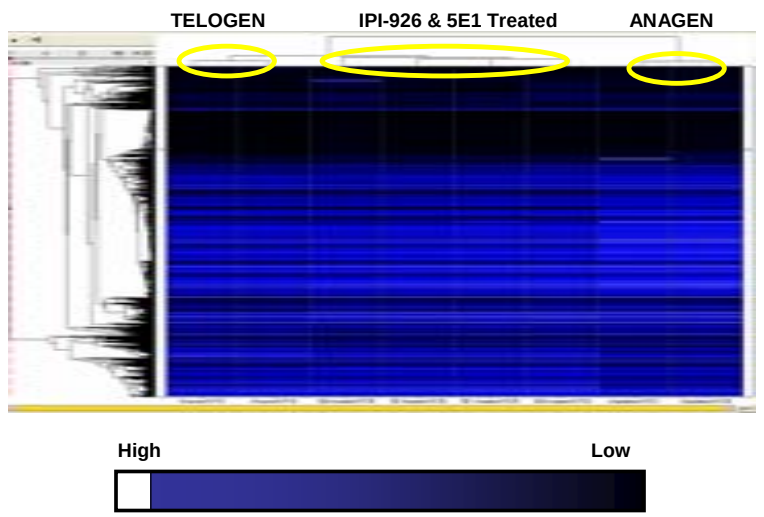

TELOGEN
IPI-926 & 5E1 Treated
ANAGEN
High
Low

To identify a group of genes that are modulated upon Hedgehog pathway inhibition, RNA from telogen, anagen as well as both IPI-926 and 5E1 treated skin was sent for Agilent array analysis. A "Hedgehog Gene Signature" was identified and its utility in pre-clinical samples is under investigation.

SUMMARY

Hedgehog target genes, including Gli-1 are induced in skin and/or hair follicles during anagen.

IPI-926 can significantly inhibit anagen induced Hedgehog target gene expression in a dose proportional manner.

Sustained Hedgehog pathway inhibition prevents anagen initiation and hair regrowth post depilation.

Measurement of the Hedgehog gene signature in the skin and/or plucked hair follicles offers an attractive and obtainable biomarker for Hedgehog antagonists in the clinic.